Safety and effectiveness of plasmapheresis-based elimination of soluble TNFα receptors combined with chemotherapy in advanced, chemorefractory triple-negative breast cancer patients - a phase I/II study (CP7-005) ***NCT04004910***

Piotr J. Wysocki[1], Adam Ostrowski[2], Robert Segal[2], Victoria Manax[2], Pawel Potocki[1], Kamil Konopka[1], Lukasz Kwinta[1], Paulina Fraczek[1], Maciej Lubas[1], Mateusz Lobacz[1], Robert Chrzan[1]. Lawrence Florin[2]

[1]Jagiellonian University Medical College Hospital, Cracow, Poland, [2]Immunicom Inc, San Diego, CA

INTRODUCTION

Tumor necrosis factor-alpha (TNFα) is a pleiotropic cytokine with known antitumor activity, produced mainly by activated immune cells. Cancer cells neutralize TNFα by shedding soluble TNF receptors 1&2 (sTNF-Rs), which act as TNFα-binding decoys, promoting cancer cell proliferation, survival, and chemoresistance (Fig 1., Tabl. 1). Immunopheresis® employs therapeutic apheresis with the selective immunoadsorption LW-02 column (LW-02) for treating solid malignancies. LW-02-based Immunopheresis® (granted FDA Breakthrough Device Designation and a CE Mark for mTNBC) selectively removes sTNF-Rs from plasma, permitting TNFα to bind to membrane-bound TNF-Rs, activating intracellular death pathways, and also modulate T-cell-mediated immune activity. Part A data of our phase I/II clinical trial in metastatic, chemorefractory, triple-negative breast cancer (mTNBC) patients (NCT04004910) confirmed that LW-02-based Immunopheresis monotherapy is safe and well-tolerated, with signs of disease stabilization (SD) in patients treated >4 weeks. Here we present interim data on the safety and preliminary efficacy of LW-02 Immunopheresis combined with chemotherapy.

CONTACT

Piotr J. Wysocki MD, PhD
Jagiellonian University Medical College Hospital
piotr.wysocki@uj.edu.pl

RATIONALE

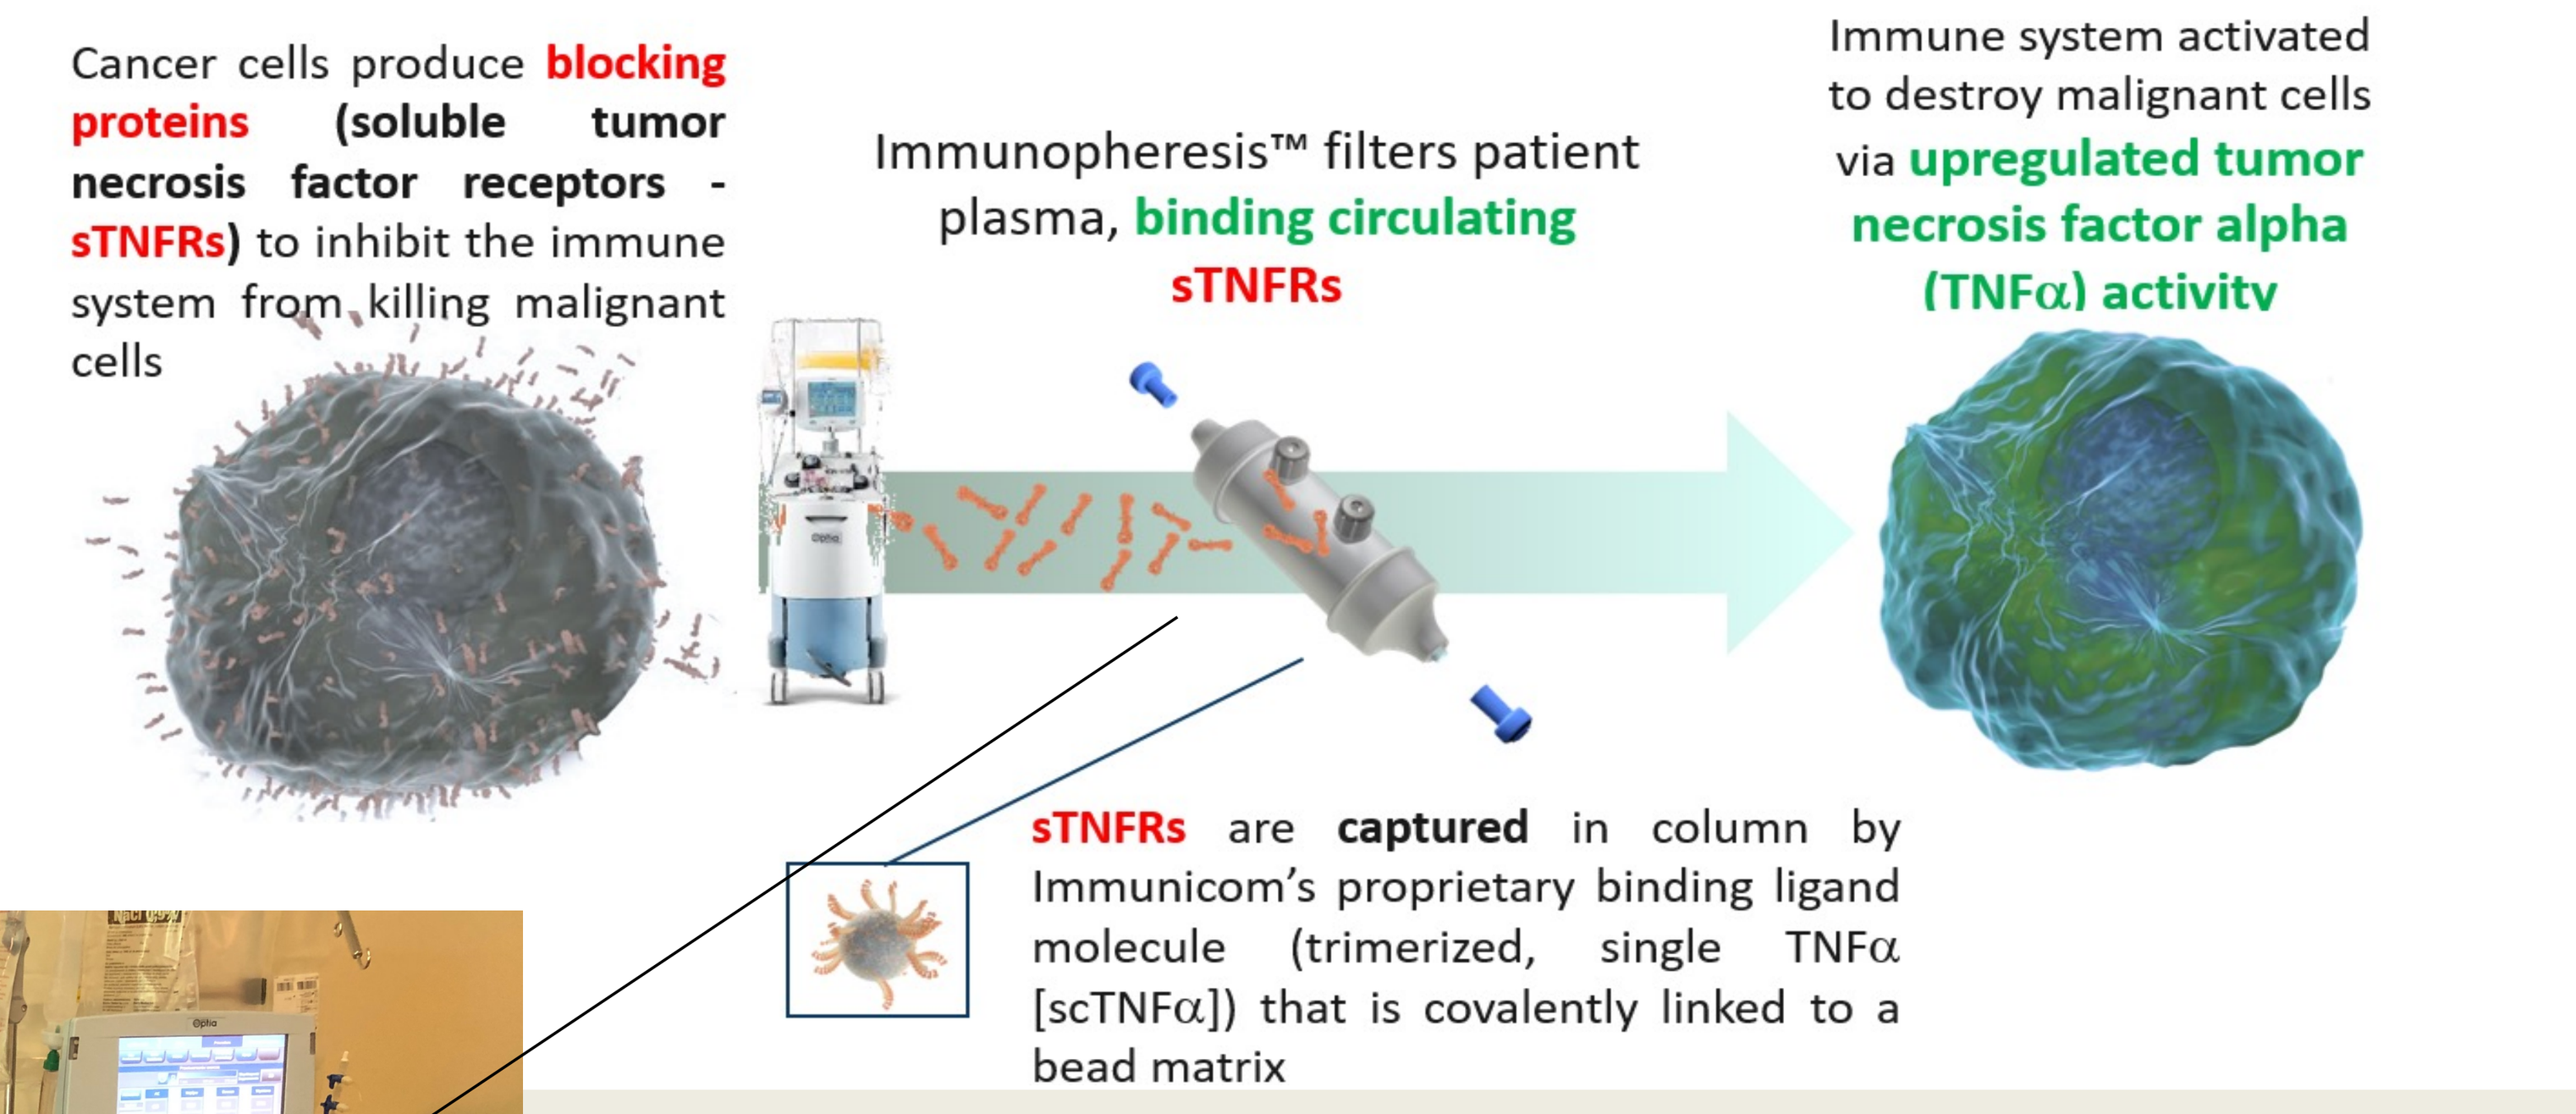
Cancer cells produce blocking proteins (soluble tumor necrosis factor receptors - sTNFRs) to inhibit the immune system from killing malignant cells
Immunopheresis™ filters patient plasma, binding circulating sTNFRs
Immune system activated to destroy malignant cells via upregulated tumor necrosis factor alpha (TNFα) activity
sTNFRs are captured in column by Immunicom's proprietary binding ligand molecule (trimerized, single TNFα [scTNFα]) that is covalently linked to a bead matrix

Figure 1. General concept

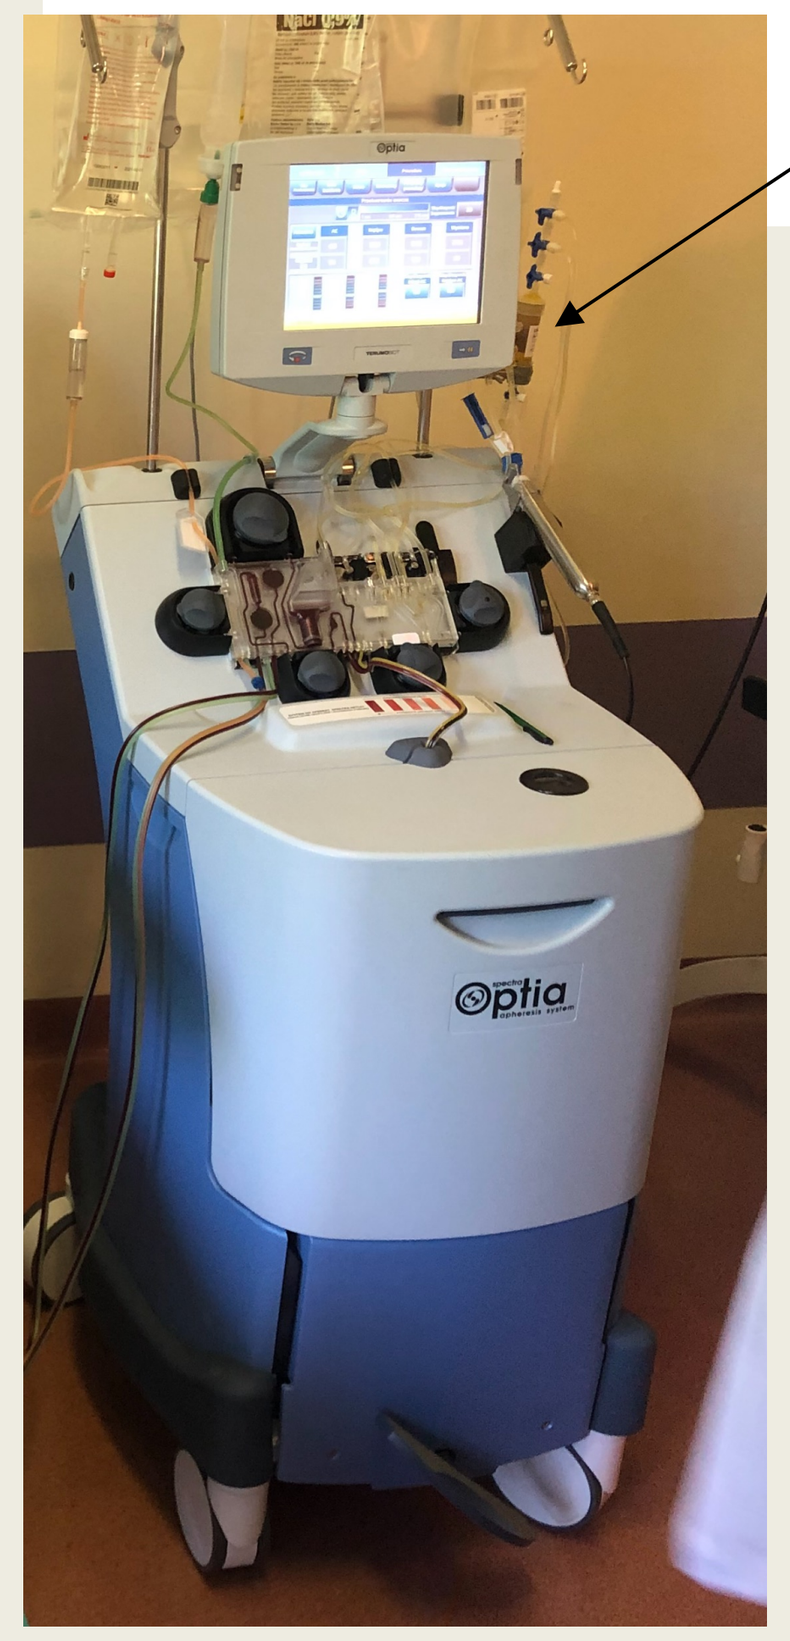

Figure 2. Spectra Optia

LW-02 – MECHANISMS OF ACTION

TNFα – related
- tumor cell apoptosis
- microvascular collapse
- stimulation of M1 macrophages
- inhibition of Tregs

sTNFR-depletion related
- unleashing of sTNF in tumor microenvironment
- inhibition of tmTNF retrograde signaling
 - decreased survival
 - decreased chemoresistance

Table 1. Serum concentrations of sTNFR1&2 in various tumors

Values (pg/ml)		Melanoma N=45	Sarcoma N=43	NSCLC N=40	RCC N=31	Ovarian N=42	Breast N=30	Normal N=20
sTNF-R1 (expressed on tumor cells)	Mean	**1,964 ±462**	**1,502 ±356**	**1,554 ±311**	**1,865 ±437**	**1,549 ±355**	**1,478 ±349**	**670 ±232**
	Range	934 - 2,886	723 - 2,136	908 - 2,212	554 - 2661	866 - 2,420	1,030 - 2,665	75 - 1,285
sTNF-R2 (expressed on immune cells)	Mean	**1,537 ±312**	**1,259 ±315**	**1,351 ±291**	**1,490 ±282**	**1,359 ±345**	**1,446 ±231**	**568 ±204**
	Range	1,054 - 2,146	476 - 1,767	610 - 1,775	1,030 - 1,989	509 - 1,872	852 - 2,108	74 - 1,094

TREATMENT

LW-02 Immunopheresis® (3x/week for 16 weeks) using a Spectra Optia device (fig. 2) is combined with paclitaxel (80 mg/m2) + carboplatin (AUC 2) d 1,8 q3w [part B] or irinotecan(125 mg/m2)+gemcitabine(750 mg/m2) d1,8 q3w [Part B-ext] in patients with mTNBC. Treatment lasts 16 weeks in clinical and/or objective responders. Primary endpoints are safety and tolerability. Secondary endpoints assessed in patients treated >4 weeks include overall survival, tumor response according to RECIST, rate of CNS progression and quality-of-life.

Of 11 patients enrolled in Part B of the study, 7 patients were treated for >4 weeks. The ORR was 18% (1CR, 1PR) with one additional patient experiencing SD; the ORR and CBR in patients treated >4 weeks were 29% and 43%, respectively. The median OS, to date, is 18.6 weeks and 26.7 weeks in the group treated >4 weeks. The rate of CNS progression (new or preexisting lesions) appears lower than expected. The most common adverse events (AEs) were chemotherapy-induced myelotoxicity (anemia, neutropenia, thrombocytopenia) and transient electrolyte abnormalities. Thirty-one serious AEs (SAEs) were reported for all 11 patients, 15 (48.4%) were transfusion requiring anemias. Of the SAEs, 3 (9.6%) were judged to have a potential causal relationship to LW-02 column Immunopheresis®.

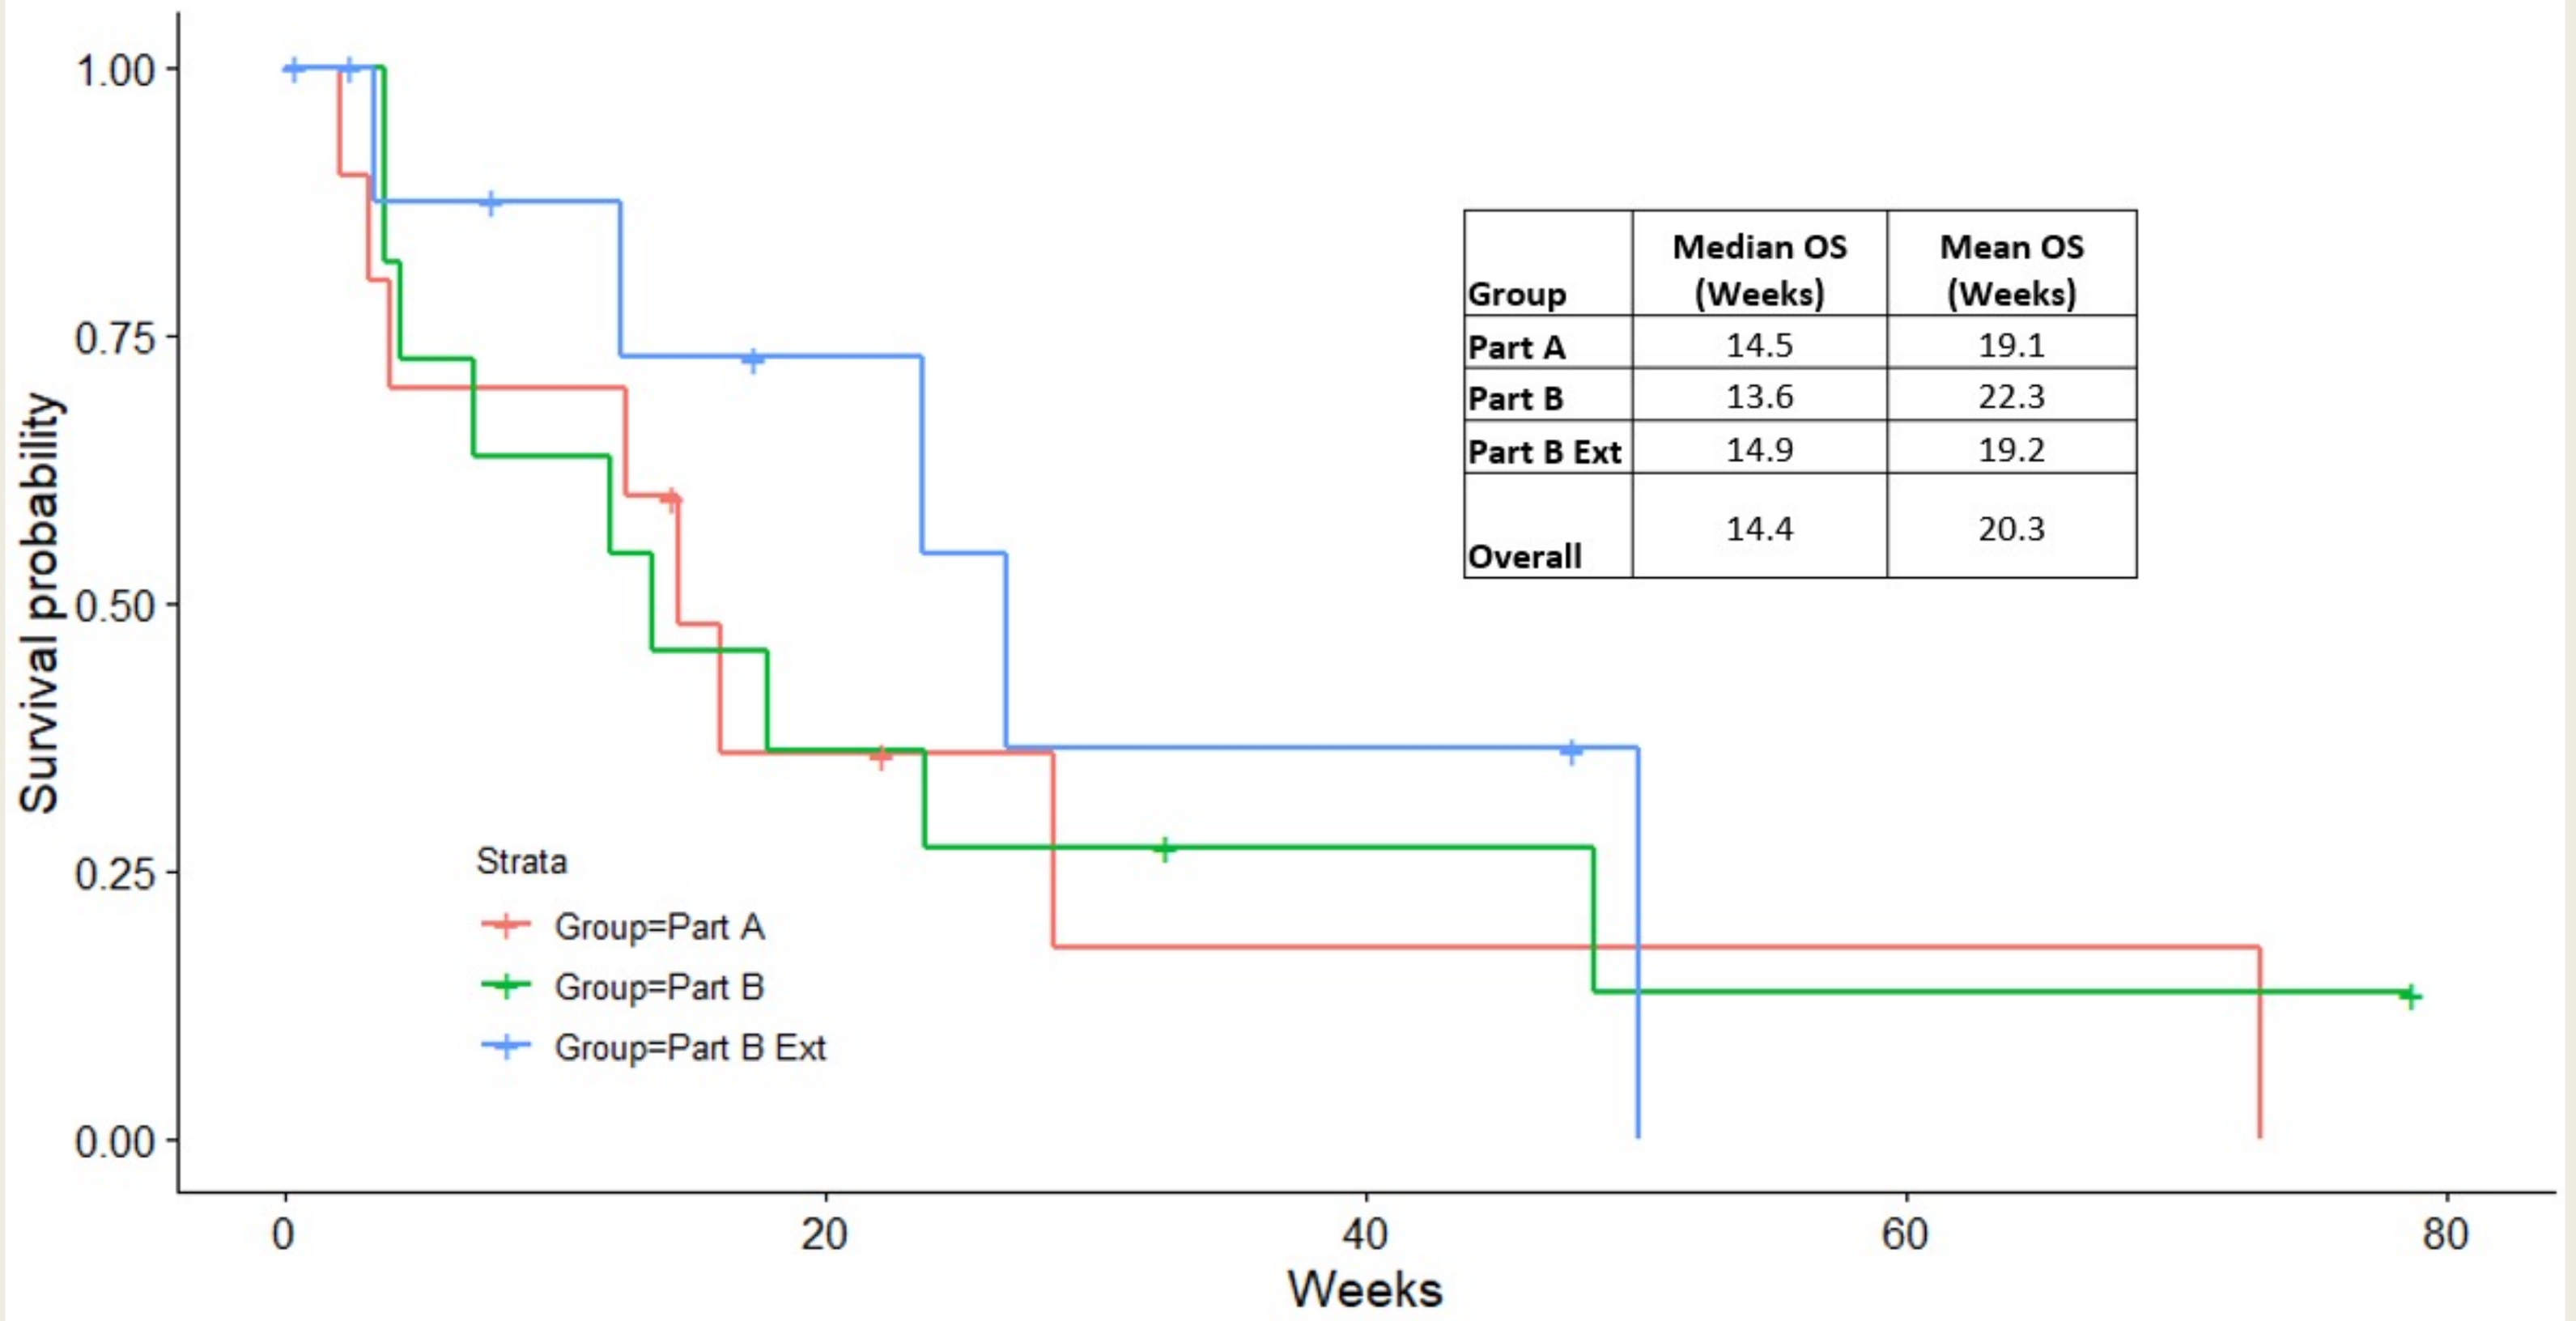
Survival probability
Weeks
Strata
Group=Part A
Group=Part B
Group=Part B Ext
Group | Median OS (Weeks) | Mean OS (Weeks)
Part A | 14.5 | 19.1
Part B | 13.6 | 22.3
Part B Ext | 14.9 | 19.2
Overall | 14.4 | 20.3

Figure 3. Overall survival of all patients in CP7-005 study. Part A: LW-02-based Immunopheresis alone, Part B: LW-02 + chemotherapy (paclitaxel + carboplatin), Part B-ext: LW-02 + chemotherapy (irinotecan + gemcitabine)

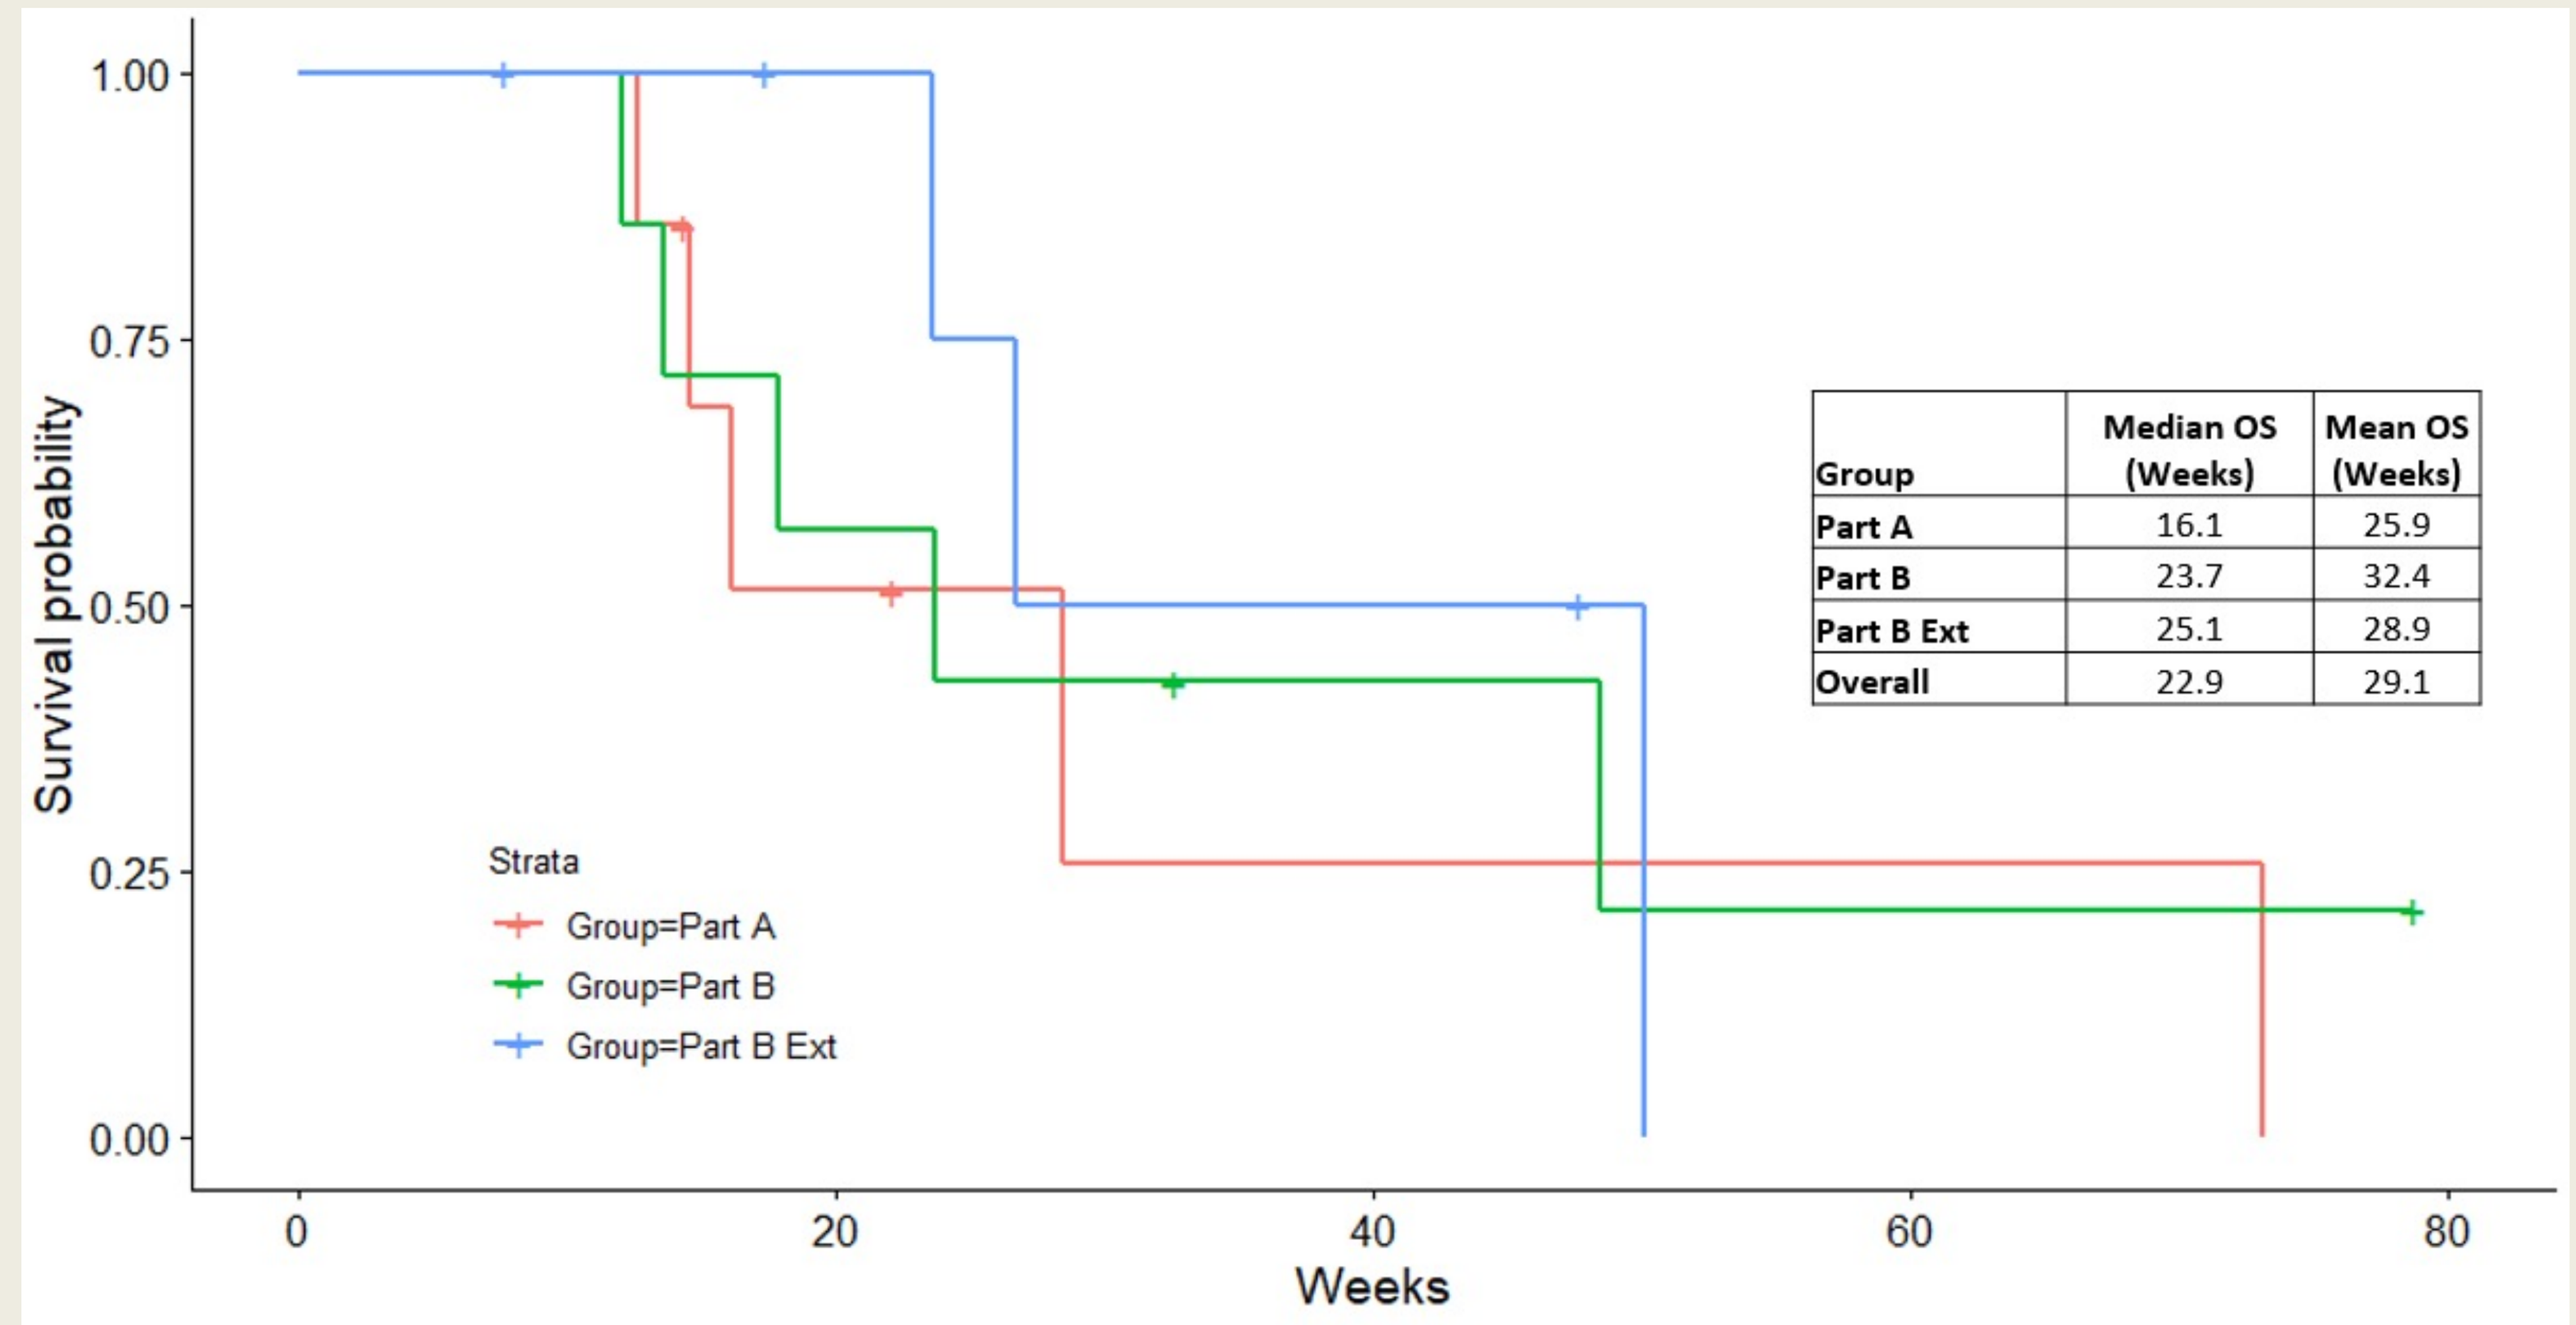
Survival probability
Weeks
Strata
Group=Part A
Group=Part B
Group=Part B Ext
Group | Median OS (Weeks) | Mean OS (Weeks)
Part A | 16.1 | 25.9
Part B | 23.7 | 32.4
Part B Ext | 25.1 | 28.9
Overall | 22.9 | 29.1

Figure 4. Overall survival of patients in CP7-005 study treated >4 weeks. Part A: LW-02-based Immunopheresis alone, Part B: LW-02 + chemotherapy (paclitaxel + carboplatin), Part B-ext: LW-02 + chemotherapy (irinotecan + gemcitabine)

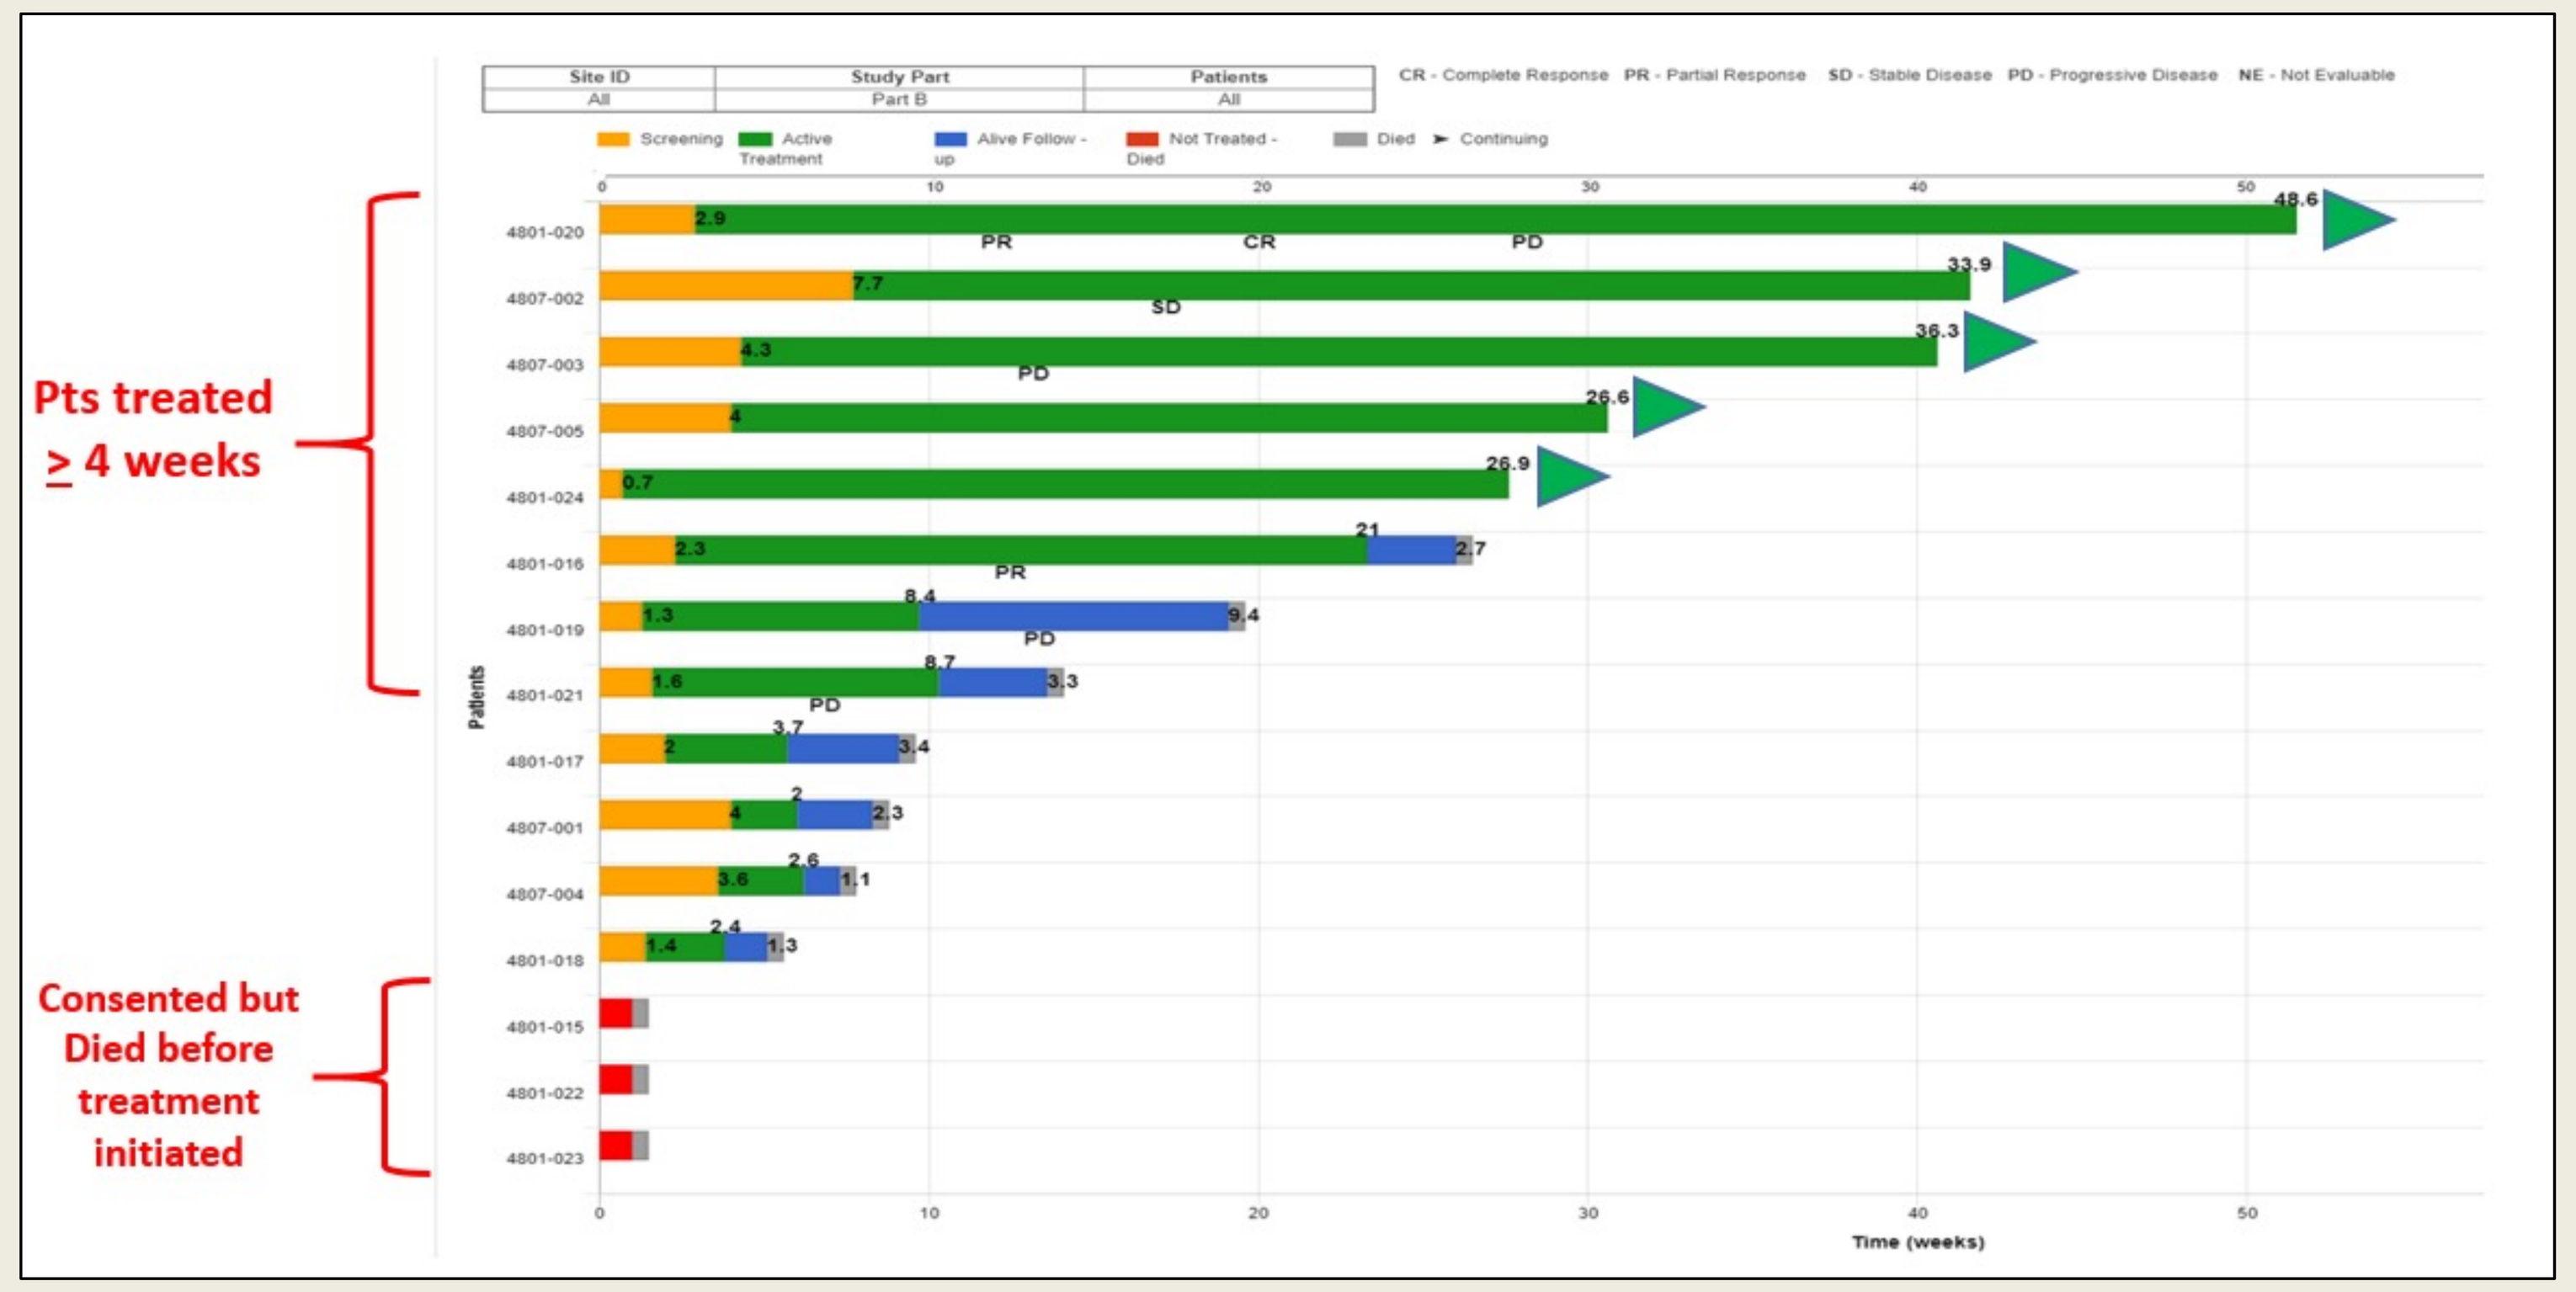
Pts treated ≥ 4 weeks
Consented but Died before treatment initiated
Patients
Time (weeks)

Figure 5. Swimmer plot of patients treated with LW-02-based Immunopheresis + chemotherapy

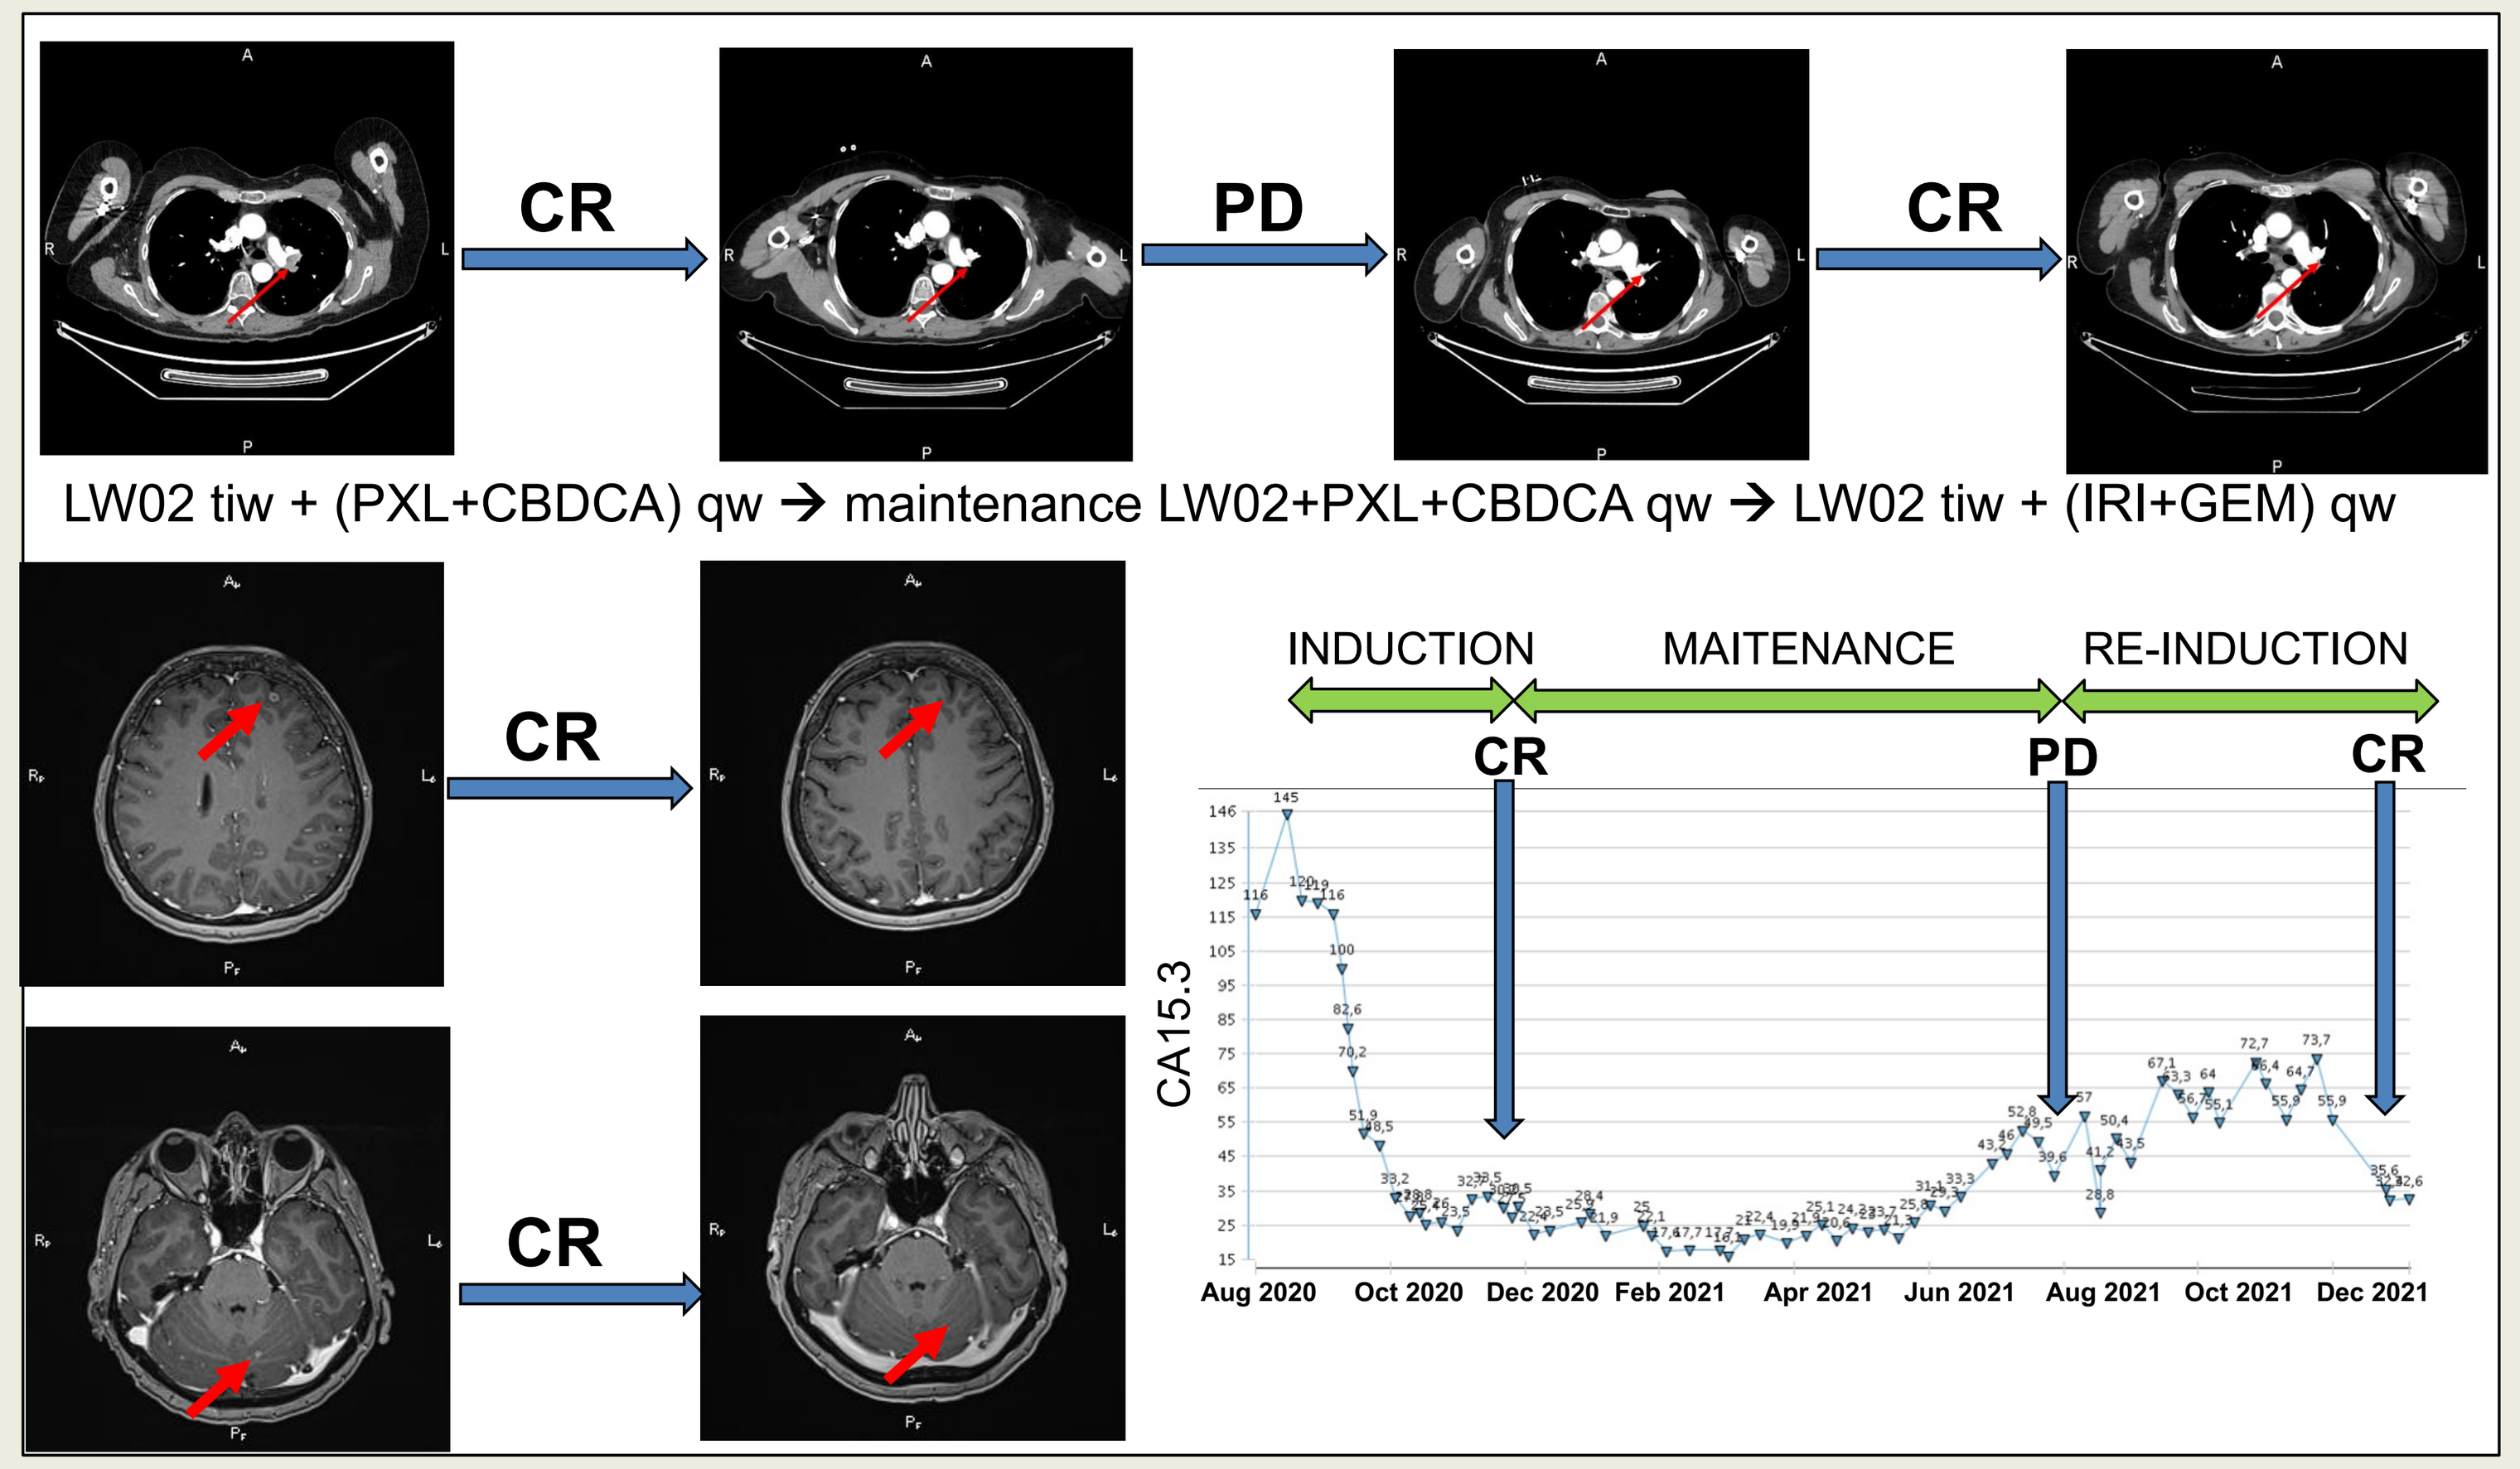
CR
PD
CR
LW02 tiw + (PXL+CBDCA) qw → maintenance LW02+PXL+CBDCA qw → LW02 tiw + (IRI+GEM) qw
CR
CR
INDUCTION
MAITENANCE
RE-INDUCTION
CR
PD
CR
CA15.3
Aug 2020
Oct 2020
Dec 2020
Feb 2021
Apr 2021
Jun 2021
Aug 2021
Oct 2021
Dec 2021

Figure 6. Presentation of a patient (after 3 lines of previous chemotherapy) with complete response (CR) to LW-02 + carboplatin + paclitaxel after 4 months of treatment, with progression (PD) after 9 months of maintenance, and subsequent CR (after 5 months) to LW-02 + irinotecan + gemcitabine

CONCLUSIONS

LW-02-based Immunopheresis® combined with weekly chemotherapy is generally safe, well-tolerated and highly effective in specific sTNFR subtraction on a longer-term basis, with promising signals of clinical benefit in heavily pretreated mTNBC patients (median 3.3 [2-5] prior lines of systemic therapy). Further clinical evaluation of the antitumor activity of LW-02-based Immunopheresis combined with low-dose chemotherapy is ongoing with a focus on quality-of-life and prevention of CNS disease progression, the latter especially important in chemorefractory mTNBC, given the high prevalence of CNS involvement.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com